Phase 2 PIONEER Trial of Oral T3D-959 for the Treatment of Patients Diagnosed with Mild-to-Moderate Alzheimer's Disease: New Results in a Modified Intent-to-Treat Population

Sunday-798

Jessica Stanek, PhD, Blake Swearingen, MS, Warren Strittmatter, MD, Stan Chamberlain, PhD, Hoda Gabriel, PMP, Christopher Coutlee, PhD, Charles Lineberry, PhD and **John Didsbury, PhD**, T3D Therapeutics, Inc., Research Triangle Park, NC, USA

- **mITT ("AD") population study results show efficacy trends and statistical significance on multiple AD pathologies with low Ns of ~30 patients/treatment group**
- **Clinical evidence of slowing in cognitive decline, by 73% in 24 weeks, a benefit of 1.97 points on ADAS-Cog11**
- **Clinical evidence of a statistically significant reduction in amyloid plaque burden comparable to anti-amyloid plaque antibodies but without clinical evidence of ARIA and once daily oral dosing without infusions**
- **Clinical evidence of a potential safety advantage over marketed therapies**
- **Biomarker results show T3D-959 to affect all three AD diagnostic criteria Amyloid/Tau/ Neurodegeneration (A/T/N) and more, including inflammation, insulin resistance and dysfunctional metabolism**
- **Potential for disease-modifying activities demonstrated. Multiple biomarkers of disease pathologies positively changed by drug, including those associated with amyloid plaque burden**
- **mITT population study results strongly support progression to Phase 3 in mild to moderate AD patients assessing a 30mg dose**

INTRODUCTION

PIONEER was a Phase 2 randomized, placebo-controlled, multi-center trial evaluating efficacy and safety of the dual PPARδ/γ agonist T3D-959 in patients with mild-to-moderate Alzheimer's disease (AD). 250 subjects were randomized in the Intent-to-Treat (ITT) population and dosed orally q.d. x 24-week at 36 US clinical trial sites.

Three significant data irregularities, discovered after unblinding, were noted in a cluster of 5 sites ("excluded sites"):

1) Pharmacokinetic anomalies: Fifty-five percent of drug-assigned subjects at the excluded sites were not taking drug, as assessed by PK and confirmed by other sensitive biomarkers.

2) Scientifically improbable biological diagnosis of Alzheimer's disease (AD): Greater than 75% of excluded site subjects did not meet the threshold for AD pathology at baseline, as assessed by validated biomarker p-tau217/np-tau217 ratio (C_2N Diagnostics, LLC) while greater than 75% of mITT did meet the threshold for AD pathology.

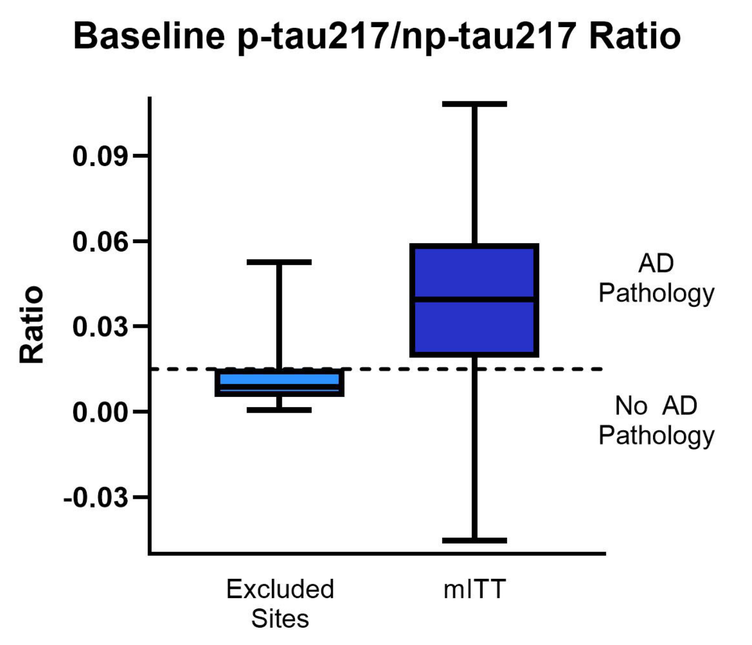

Baseline p-tau217/np-tau217 Ratio
Ratio
0.09
0.06
0.03
0.00
-0.03
AD Pathology
No AD Pathology
Excluded Sites
mITT

3) Scientifically improbable Placebo Response: Subjects' placebo response on primary endpoints at excluded sites was well outside of historic norms and inconsistent with the diagnosis of AD.

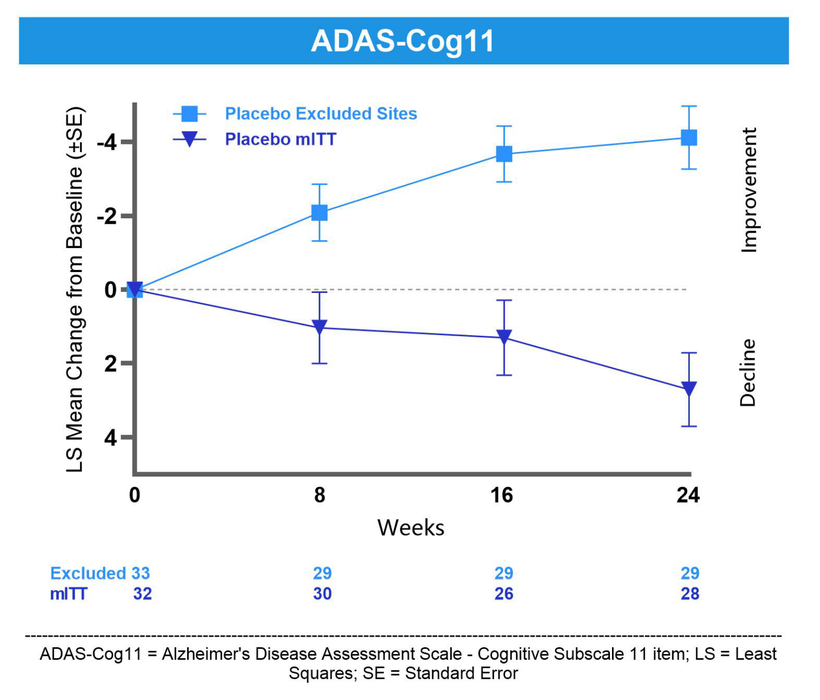

ADAS-Cog11
Placebo Excluded Sites
Placebo mITT
LS Mean Change from Baseline (±SE)
Improvement
Decline
Weeks
Excluded 33 29 29 29
mITT 32 30 26 28

ADAS-Cog11 = Alzheimer's Disease Assessment Scale - Cognitive Subscale 11 item; LS = Least Squares; SE = Standard Error

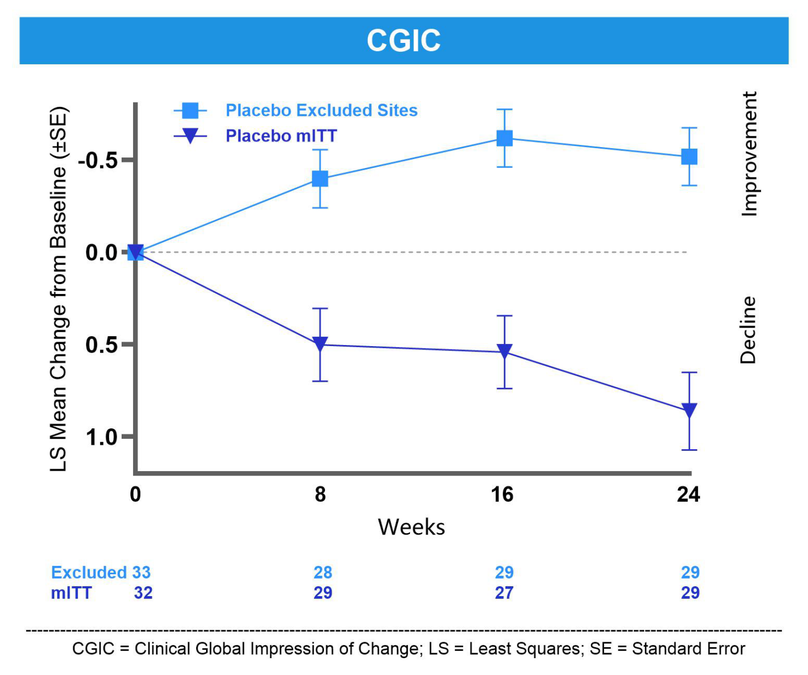

CGIC
Placebo Excluded Sites
Placebo mITT
LS Mean Change from Baseline (±SE)
Improvement
Decline
Weeks
Excluded 33 28 29 29
mITT 32 29 27 29

CGIC = Clinical Global Impression of Change; LS = Least Squares; SE = Standard Error

Developing innovative AD therapies, especially those that can be orally administered, comprise a significant public health imperative. While the original intent-to-treat PIONEER analysis failed on its primary endpoint, positive effects indicative of significant T3D-959 therapeutic benefit have been identified across a spectrum of study endpoints. The inclusion of data from the 5 unevaluable study sites confounded our ability to evaluate therapeutic effect accurately and completely in the PIONEER study. The ITT population was modified to remove data from these 5 sites, defining a Modified Intent-to-Treat (mITT) population of 141 subjects. The mITT population was also independently corroborated using blinded study data (Pentara Corp.) and provides an interpretable dataset with high data integrity. Per formal discussion with the FDA, the mITT will be reported in the Clinical Study Report (CSR) as a post-hoc efficacy analysis.

METHODS

In the PIONEER Study 250 patients were randomized to placebo (n=65) or T3D-959 15-mg, (n=63); 30-mg, (n=62); or 45-mg, (n=60). Enrolled patients had mild-to-moderate AD per NIA-AA criteria. No AD biomarker enrollment criteria were defined. Data from all subjects enrolled at the five excluded sites were removed from the dataset to form an mITT study population of 141 patients. Primary endpoints were ADAS-Cog11 and ADCS-CGIC. Secondary endpoints were plasma Aβ42/40 ratio and Digit Symbol Coding Test. Proteomic biomarkers including plasma p-tau217 ratio (C_2N) and plasma neurogranin (Inoviv) were exploratory endpoints. Efficacy was evaluated as least-squares mean changes from baseline to week 24.

RESULTS

A. Subject Disposition

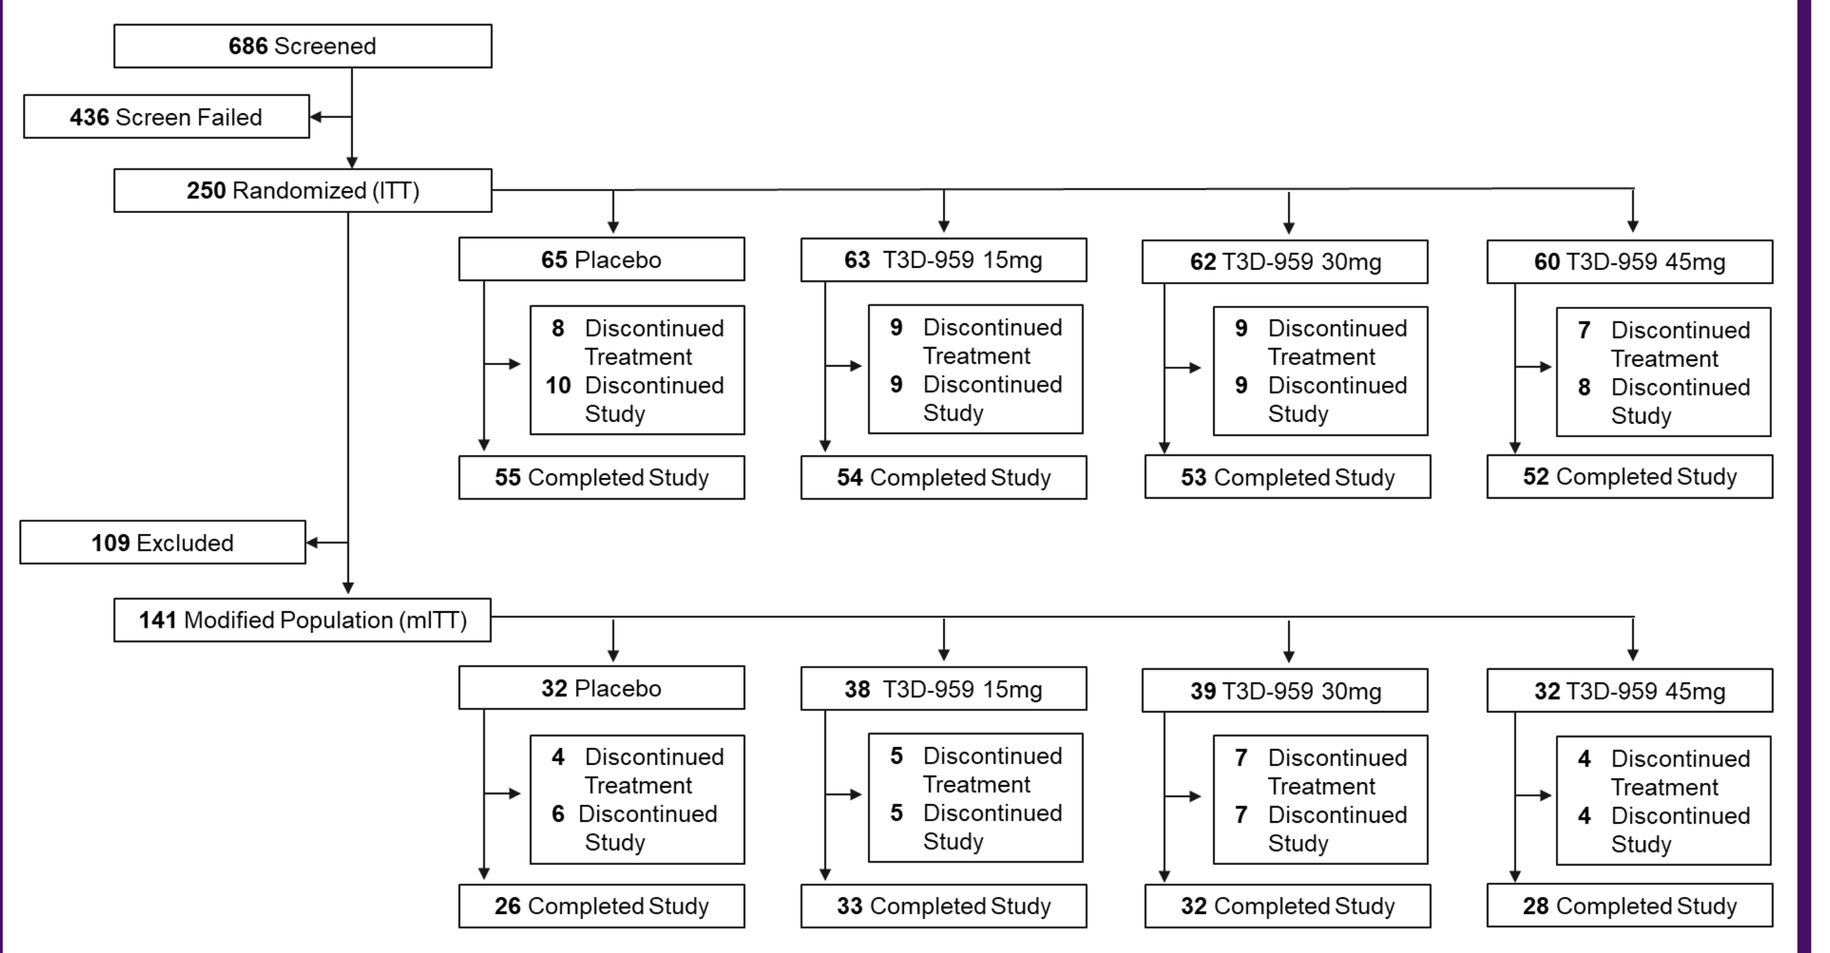

686 Screened
436 Screen Failed
250 Randomized (ITT)
65 Placebo
8 Discontinued Treatment
10 Discontinued Study
55 Completed Study
63 T3D-959 15mg
9 Discontinued Treatment
9 Discontinued Study
54 Completed Study
62 T3D-959 30mg
9 Discontinued Treatment
9 Discontinued Study
53 Completed Study
60 T3D-959 45mg
7 Discontinued Treatment
8 Discontinued Study
52 Completed Study
109 Excluded
141 Modified Population (mITT)
32 Placebo
4 Discontinued Treatment
6 Discontinued Study
26 Completed Study
38 T3D-959 15mg
5 Discontinued Treatment
5 Discontinued Study
33 Completed Study
39 T3D-959 30mg
7 Discontinued Treatment
7 Discontinued Study
32 Completed Study
32 T3D-959 45mg
4 Discontinued Treatment
4 Discontinued Study
28 Completed Study

B. Baseline Characteristics – mITT vs Excluded Sites

Baseline characteristics of mITT population is consistent with the clinical and biological diagnosis of mild-to-moderate AD.

	Excluded Sites	mITT
Enrolled	109 (43.6%)	141 (56.4%)
Female	82 (75.2%)	74 (52.5%)
Age	70.9 (± 7.43)	74.4 (± 8.08)
Hispanic/Latino	108 (99.1%)	22 (15.6%)
Education <= 12 years	74 (67.9%)	33 (23.4%)
MMSE	21.3 (± 2.70)	20.0 (± 3.60)
ADAS-Cog11	17.1 (± 5.27)	21.3 (± 8.58)
APOE4 Positive	20 (18.3%)	86 (61.0%)
Amyloid Probability Score High	30 (27.5%)	107 (77.5%)
Concomitant AD Therapy	71 (65.1%)	94 (66.7%)
HbA1c Prediabetes	70 (70.0%)	74 (52.2%)
p-tau217/np-tau217 Ratio	0.0117 (± 0.00962)	0.0409 (± 0.02643)

Continuous variables are mean ± standard deviation

C. Co-Primary Endpoint – ADAS-Cog11

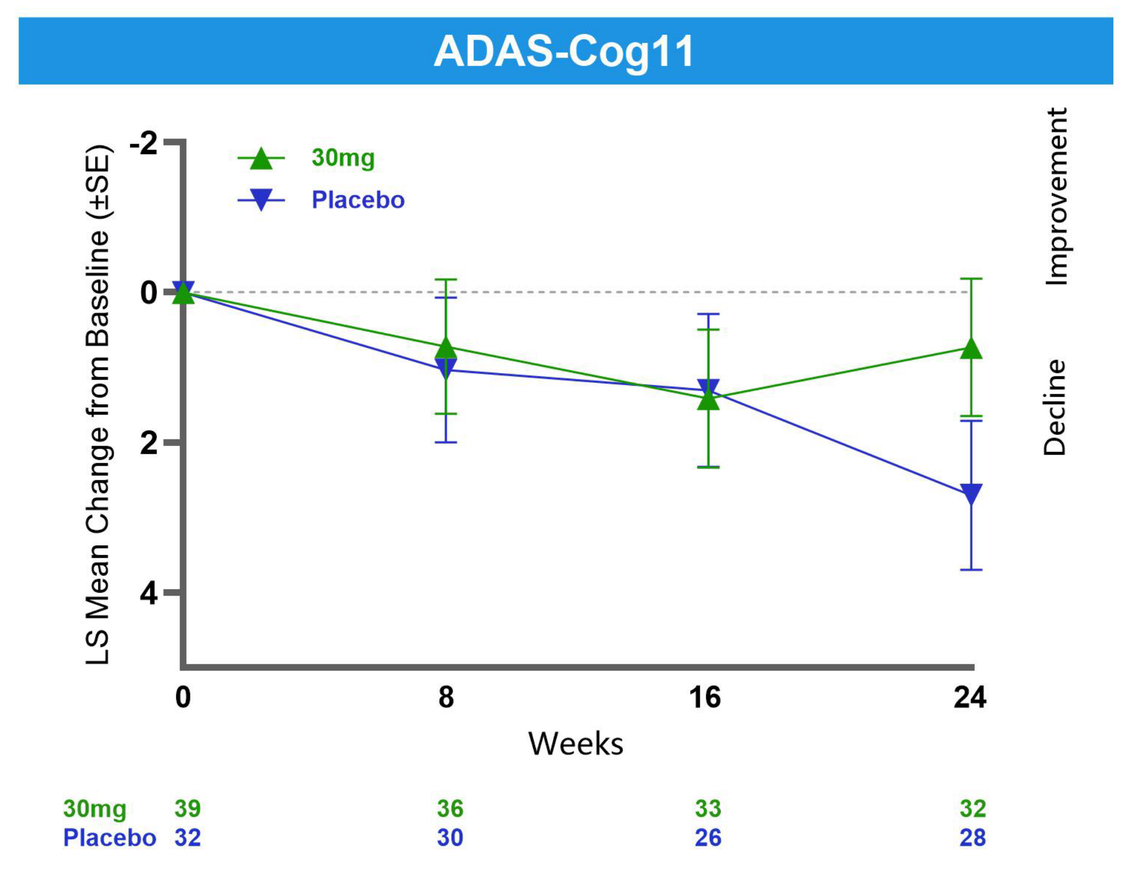

ADAS-Cog11
30mg
Placebo
LS Mean Change from Baseline (±SE)
Improvement
Decline
Weeks
30mg 39 36 33 32
Placebo 32 30 26 28

ADAS-Cog11 = Alzheimer's Disease Assessment Scale - Cognitive Subscale 11 item; LS = Least Squares; SE = Standard Error

Primary Endpoint: ADAS-Cog11

	Placebo (N=32)	T3D-959 15mg (N=38)	T3D-959 30mg (N=39)	T3D-959 45mg (N=32)
Week 8				
LS Means	1.03	0.85	0.72	3.07
SE of LS Means	0.96	0.91	0.89	0.94
p-value		0.865	0.769	0.067
Week 16				
LS Means	1.30	1.38	1.41	3.04
SE of LS Means	1.01	0.92	0.91	0.95
p-value		0.946	0.921	0.134
Week 24				
LS Means	2.70	2.39	0.73	3.81
SE of LS Means	0.99	0.92	0.91	0.98
p-value		0.778	0.073	0.337

LS = Least Squares; SE = Standard Error

- Clinical benefit of 1.97 points, corresponding to a 73% slowing in cognitive decline, after 24-wks of treatment with T3D-959 30mg vs placebo in the mITT population
- Strong trend in the mITT population, supportive for 30mg dose selection in future trials

Number (%) of Subjects with Clinically Meaningful Change ADAS-Cog11

	Placebo	T3D-959 15mg	T3D-959 30mg	T3D-959 45mg
Improvement	4 (14.3)	6 (18.2)	9 (28.1)	4 (15.4)
No Change	14 (50.0)	16 (48.5)	17 (53.1)	12 (46.2)
Worsening	10 (35.7)	11 (33.3)	6 (18.8)	10 (38.5)
P-value		0.725	0.089	0.930

P-value is based on CMH row mean scores test.
Improvement defined as a change from baseline score of -3 or less, No Change defined as change from baseline score of -2 to +2, and Worsening defined as change from baseline score of +3 or more.

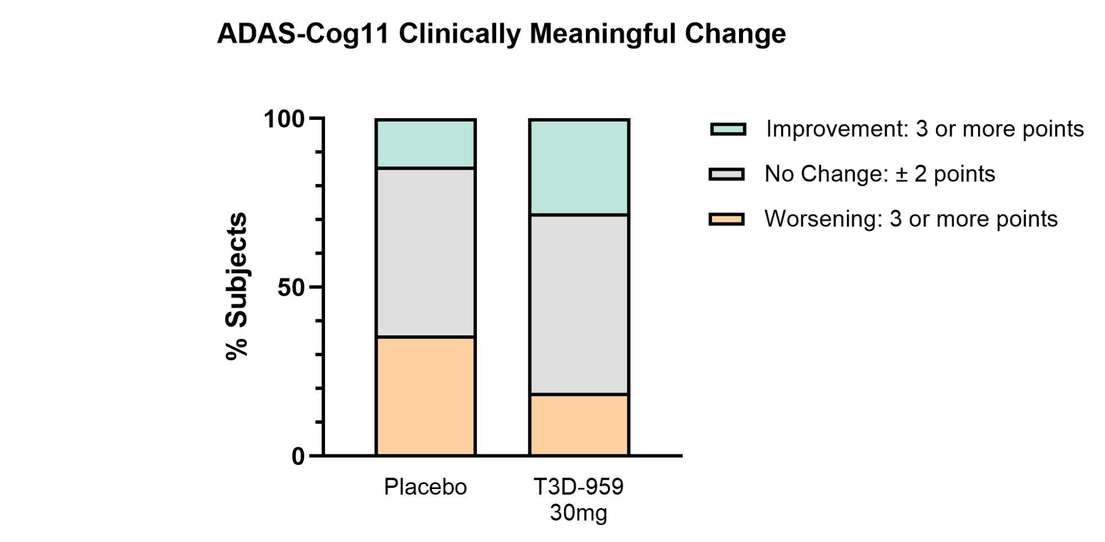

ADAS-Cog11 Clinically Meaningful Change
% Subjects
Improvement: 3 or more points
No Change: ± 2 points
Worsening: 3 or more points
Placebo
T3D-959 30mg

- T3D959 30mg dose group experienced less worsening AND more clinically meaningful* improvement than placebo (*Ref:Schrag A, Schott JM, J Neurol Neurosurg Psychiatry. 2012 Feb;83(2):171-3)

D. Co-Primary Endpoint – ADCS-CGIC

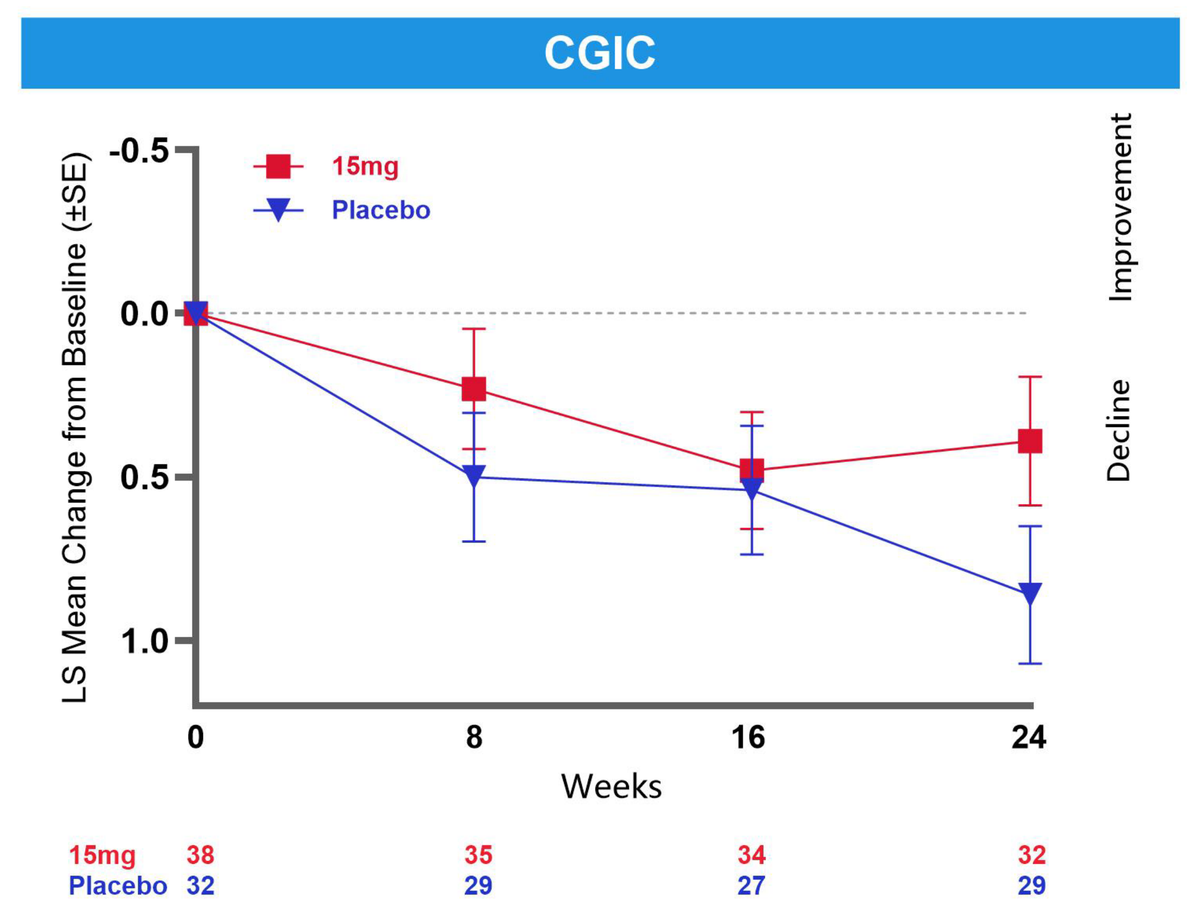

CGIC
15mg
Placebo
LS Mean Change from Baseline (±SE)
Improvement
Decline
Weeks
15mg 38 35 34 32
Placebo 32 29 27 29

CGIC = Clinical Global Impression of Change; LS = Least Squares; SE = Standard Error

Primary Endpoint: CGIC

	Placebo (N=32)	T3D-959 15mg (N=38)	T3D-959 30mg (N=39)	T3D-959 45mg (N=32)
Week 8				
LS Means	0.50	0.23	0.44	0.07
SE of LS Means	0.196	0.182	0.173	0.188
p-value		0.2372	0.796	0.0668
Week 16				
LS Means	0.54	0.48	0.73	0.42
SE of LS Means	0.196	0.178	0.172	0.189
p-value		0.7811	0.4147	0.5902
Week 24				
LS Means	0.86	0.39	0.73	0.65
SE of LS Means	0.209	0.196	0.187	0.207
p-value		0.0601	0.5994	0.4055

LS = Least Squares; SE = Standard Error

- All doses numerically improved vs placebo, with a trend for significant improvement at T3D-959 15mg vs placebo

E. Secondary Endpoint – Plasma Aβ 42/40 Biomarker

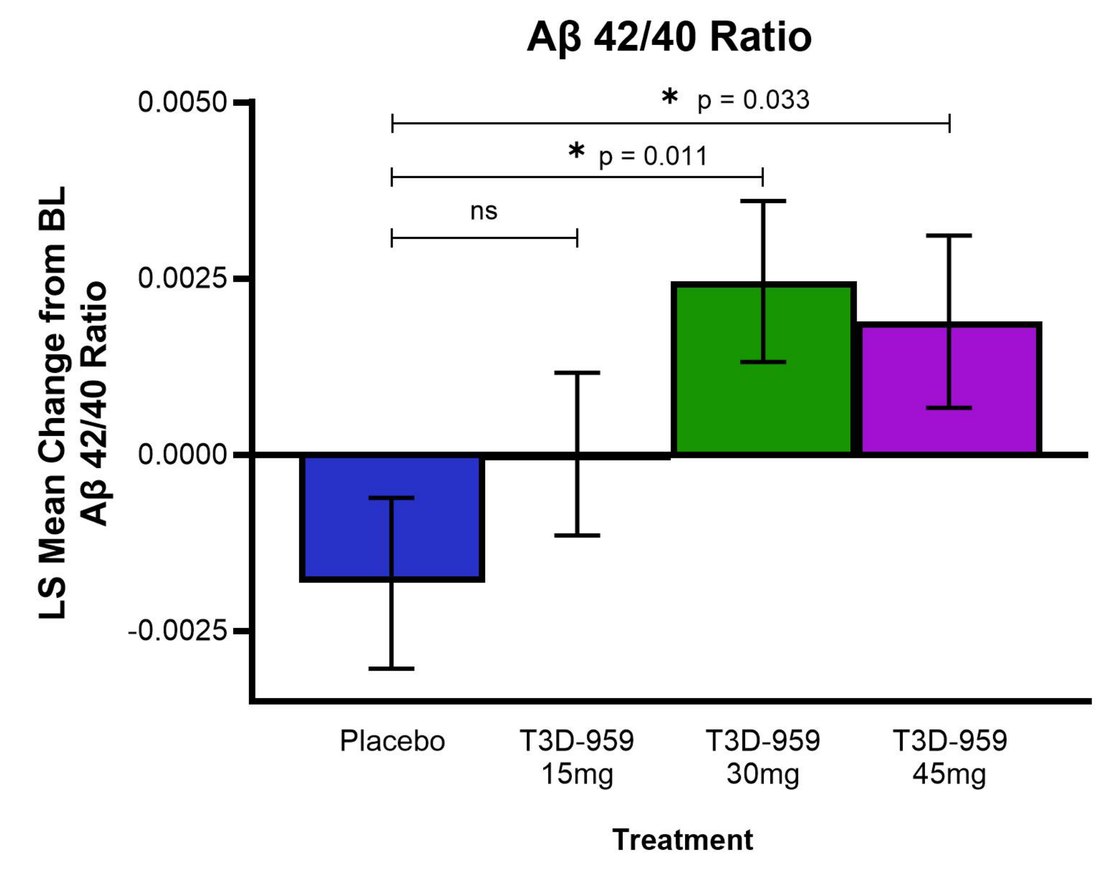

Aβ 42/40 Ratio
LS Mean Change from BL Aβ 42/40 Ratio
* p = 0.033
* p = 0.011
ns
Placebo
T3D-959 15mg
T3D-959 30mg
T3D-959 45mg
Treatment

Secondary Endpoint: Amyloid-Beta 42/40 Ratio Plasma Biomarker

	Placebo	T3D-959 15mg	T3D-959 30mg	T3D-959 45mg
Baseline				
n	32	36	39	31
Mean	0.084	0.083	0.085	0.085
SD	0.009	0.008	0.008	0.009
Week 24 EOT				
n	29	33	32	28
Mean	0.082	0.083	0.087	0.086
SD	0.007	0.006	0.009	0.008
EOT - BL				
LS means	-0.0018	0.0000	0.0025	0.0019
SE LS Means	0.0012	0.0012	0.0011	0.0012
Diff LS means		0.0018	0.0043	0.0037
p-value		0.2694	0.0108	0.0333

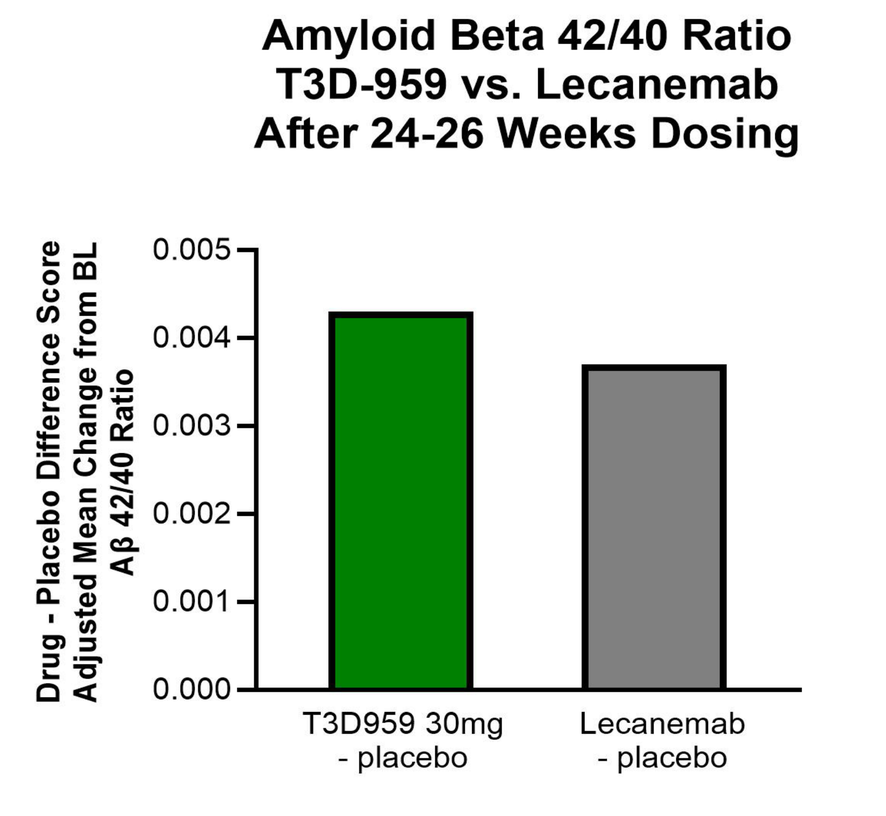

Amyloid Beta 42/40 Ratio
T3D-959 vs. Lecanemab
After 24-26 Weeks Dosing
Drug - Placebo Difference Score Adjusted Mean Change from BL Aβ 42/40 Ratio
T3D959 30mg - placebo
Lecanemab - placebo

- Statistically significant improvement in T3D-959 30mg and T3D-9595 45mg dose groups vs placebo

F. Secondary Endpoint – Digit Symbol Coding Test

Secondary Endpoint: DSCT

	Placebo (N=32)	T3D-959 15mg (N=38)	T3D-959 30mg (N=39)	T3D-959 45mg (N=32)
Week 8				
LS Means	0.43	-0.49	1.12	0.36
SE of LS Means	1.283	1.164	1.101	1.216
p-value		0.5251	0.6406	0.9649
Week 16				
LS Means	-2.03	0.07	-1.74	-0.08
SE of LS Means	1.648	1.444	1.423	1.528
p-value		0.2901	0.8864	0.3535
Week 24				
LS Means	-2.66	-0.93	-1.33	0.55
SE of LS Means	1.564	1.411	1.403	1.512
p-value		0.362	0.4933	0.1124

LS = Least Squares; SE = Standard Error

- All doses numerically improved vs placebo, with a trend for significant improvement in T3D-959 45mg vs placebo

G. Exploratory Endpoint – ("A/T" Biomarker): Plasma p-tau217/non-p-tau217 Ratio

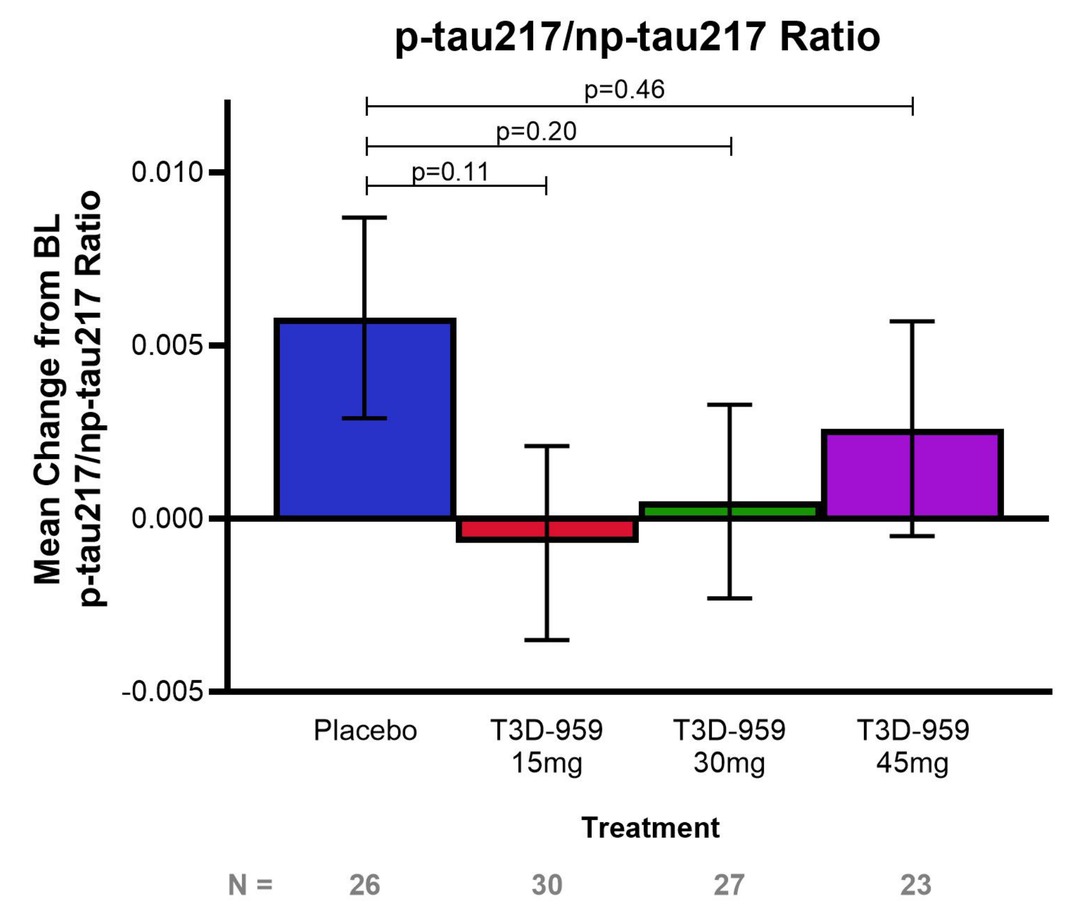

p-tau217/np-tau217 Ratio
Mean Change from BL p-tau217/np-tau217 Ratio
p=0.46
p=0.20
p=0.11
Placebo
T3D-959 15mg
T3D-959 30mg
T3D-959 45mg
Treatment
N = 26 30 27 23

- Marker of AD pathology, associated with amyloid-β and tau pathologies
- Disease progression in placebo
- Disease progression slowed with drug treatment; trend at T3D-959 15mg vs placebo

H. Exploratory Endpoint – ("N" Biomarker): Plasma Neurogranin

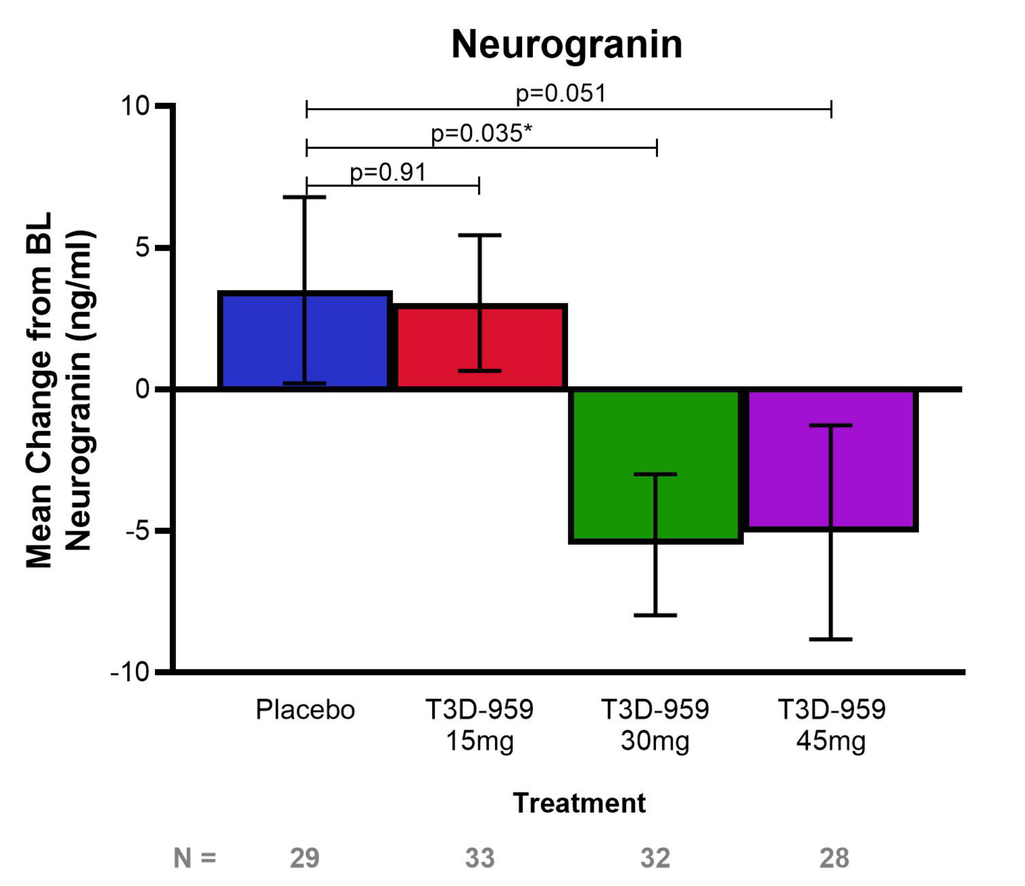

Neurogranin
Mean Change from BL Neurogranin (ng/ml)
p=0.051
p=0.035*
p=0.91
Placebo
T3D-959 15mg
T3D-959 30mg
T3D-959 45mg
Treatment
N = 29 33 32 28

- Marker of neurodegeneration increased in AD, associated with synaptic loss
- Statistically significant improvement at T3D-959 30mg and trend at T3D-959 45mg vs placebo

I. Dose Selection for Phase 3 – Greatest frequency of improvement in T3D-959 30mg dose across multiple measures

Improvement in Efficacy Measures by Dose	15mg	30mg	45mg
Neuropyschological Assessments			
ADAS-Cog11 - Primary Outcome Measure of Cognition			
ADCS-CGIC - Primary Outcome Measure of Function			
DSCT - Secondary Outcome Measure of Executive Function			
A/T/N Plasma Biomarkers			
Ab42/40 - Secondary Outcome Measure of Reduction in Amyloid Plaque Burden		*	*
p-tau217/np-tau217 Ratio - Amyloid and Tau Pathologies			
Neurogranin - Neurodegeneration, Synaptic Loss		*	
Exploratory Plasma Biomarkers			
Adiponectin - Aβ Deposition, Tau Phosphorylation, Inflammation, Insulin Resistance	*	***	*
Alpha2 Macroglobulin - Aβ Fibril Prevention, Plaque Clearance & Reducing Aβ Neurotox.			
ApoA1 - Aberrant HDL Lipid Metabolism	*		
ApoC3 - Aberrant HDL Lipid Metabolism & Inflammation			***
Ficolin 2 - Brain Atrophy, Insulin Resistance, Inflammation			
Gelsolin - Aβ Fibrilization, Oxidative Stress, Neurodegeneration			
Hemoglobin Subunit Beta - Oxidative Stress, Neurodegeneration	*	*	**
Transthyretin - Aβ Aggregation, Vascular Defects			
von Willebrand Factor - Vascular damage, White Matter Hyperintensities		*	*
Mechanism of Action (MOA) Plasma Biomarkers			
Glucose - Insulin Sensitization	*	**	
Insulin - Insulin Sensitization		**	
HOMA-IR - Insulin Sensitization		***	
Triglycerides - Dyslipidemia Related to BBB Integrity			
HDL - Dyslipidemia Related to BBB Integrity	*	**	

* = p < 0.05; ** = p < 0.01; *** = p < 0.001; All other colored cells are trending vs. placebo at p < 0.15.

J. Safety – ITT Population

- 250 subjects representing > 5,500 patient dosing weeks
- 265 AEs across 97 subjects
- 13 minor drug-related AEs and 4 placebo-related AEs across 12 subjects
- No drug-related SAEs
- 13 drug-unrelated SAEs, 1 drug-unrelated death (end stage dementia)
- 1 dropout due to drug-related AE (mild edema peripheral)
- No high frequency of any AE type

Number (%) of subjects with adverse events

	T3D-959 15mg (N=63) n (%)	T3D-959 30mg (N=62) n (%)	T3D-959 45 mg (N=60) n (%)	Placebo (N=65) n (%)
Any adverse event	21 (33.3)	26 (41.9)	22 (36.7)	28 (43.1)
Adverse event related to drug or placebo	2 (3.2)	2 (3.2)	6 (10.0)	2 (3.1)
Serious adverse event	1 (1.6)	4 (6.5)	3 (5.0)	3 (4.6)
Adverse event resulting in discontinuation	1 (1.6)	1 (1.6)	1 (1.7)	3 (4.6)
Death	1 (1.6)	0	0	0
Adverse events with incidence ≥ 3% in any group				
COVID-19	5 (7.9)	7 (11.3)	4 (6.7)	9 (13.8)
Diarrhea	2 (3.2)	4 (6.5)	3 (5.0)	2 (3.1)
Urinary tract infection	2 (3.2)	2 (3.2)	3 (5.0)	3 (4.6)
Headache	1 (1.6)	1 (1.6)	3 (5.0)	2 (3.1)
Upper respiratory tract infection	3 (4.8)	0	0	1 (1.5)
Fall	2 (3.2)	1 (1.6)	0	3 (4.6)
Oedema peripheral	0	2 (3.2)	2 (3.3)	0
Back pain	1 (1.6)	0	2 (3.3)	0
Urinary incontinence	1 (1.6)	0	0	2 (3.1)
Anaemia	0	2 (3.2)	0	0
Osteoarthritis	0	2 (3.2)	0	0

CONCLUSIONS

- Efficacy trends and statistical significance on multiple AD pathologies, particularly noteworthy given small Ns of ~30 patients/treatment group
- Clinical evidence of slowing in cognitive decline. A clinical benefit of 1.97 points, corresponding to a 73% slowing in cognitive decline, after 24-wks of treatment with 30mg q.d. dose of T3D-959
- Clinical evidence of a statistically significant reduction in amyloid plaque burden comparable to anti-amyloid plaque antibodies but without clinical evidence of ARIA and once daily oral dosing without infusions
- Clinical evidence of a potential safety advantage over marketed therapies
- Dose for Phase 3 identified
- Biomarker results show T3D-959 to affect all three AD diagnostic criteria Amyloid/Tau/Neurodegeneration (A/T/N) and more, including inflammation, insulin resistance and dysfunctional metabolism
- Potential for disease-modifying activities demonstrated. Multiple biomarkers of disease pathologies positively changed by drug, including those associated with amyloid plaque burden
- Target engagement demonstrated
- Along with the strong safety profile of T3D-959, evidence is supportive of a larger Phase 3 study evaluating T3D-959 30-mg/day in patients with mild-to-moderate AD

ACKNOWLEDGEMENTS

- Clinical National Institute on Aging, part of the National Institutes of Health, under award number R01AG061122
- The Alzheimer's Association's Part the Cloud Gates Partnership Grant Program

CONTACT

Jessica Stanek, PhD
VP Clinical Development
jessicastanek@t3dtherapeutics.com
919-237-4897

ALZHEIMER'S ASSOCIATION
AAIC 24
Alzheimer's Association International Conference

The views and opinions expressed do not necessarily reflect the Alzheimer's Association.